Benefits of Surgical Implantation of an Investigational Biodegradable Neuro-Spinal Scaffold in Various Animal Models of Acute Spinal Cord Contusion Injury: Clinical Translation

Alex A. Aimetti, Ph.D.[1], James D. Guest, M.D., Ph.D.[2], Nicholas Theodore, M.D.[3], Domagoj Coric, M.D.[4], Lorianne Masuoka, M.D.[1], Simon W. Moore, Ph.D.[1], Richard T. Layer, Ph.D.[1], Thomas R. Ulich, M.D.[1]

[1]InVivo Therapeutics Corporation, Cambridge, MA, USA [2]Department of Neurosurgery, University of Miami, Miami, FL, USA [3]Division of Neurological Surgery, Barrow Neurological Institute, St. Joseph's Hospital and Medical Center, Phoenix, AZ, USA [4]Department of Neurosurgery, Carolinas Medical Center, Carolina Neurosurgery and Spine Associates, Charlotte, NC, USA

Introduction

The pathophysiological sequelae of primary spinal cord injury (SCI) include edema, spinal cord swelling, reduced blood flow, and local tissue ischemia resulting in further cellular necrosis culminating in the appearance of a tissue void (cavity). Biodegradable scaffolds can be implanted within the necrotic lesion to fill this void and provide structural support to the surrounding viable tissue while serving as a locus for appositional healing.[1,2] Here we present results of Neuro-Spinal Scaffold implantation into the contused spinal cord of rats and pigs, as well as a clinical update on our safety and feasibility study of Neuro-Spinal Scaffold implantation into acute, neurologically complete (AIS A) spinal cord injured patients.

Methods

Neuro-Spinal Scaffold Fabrication

PLGA (50:50) with a terminal acid functional group was reacted to poly(L-lysine) (PLL) similar to a previously published method.[3] Cylindrical, porous PLGA-PLL implants were then fabricated using a solvent casting – porogen leaching (SCPL) process as described previously.[1] Scaffolds were terminally sterilized using electron beam radiation.

Porcine Contusion Spinal Cord Injury Model – Surgical Feasibility

Gottigen pigs (*n=4)* were contused at T10 with a 50 g weight dropped from 40 cm, followed by a 100 g compression for 5 minutes as described previously.[4] At 4, 6 and 24 hours following injury, the necrotic injury center was gently debrided following myelotomy and the Neuro-Spinal Scaffold was implanted. Intraparenchymal pressure measurements were obtained during the surgical implantation at 24 hours post-injury.

Rat Contusion Spinal Cord Injury Model – Preservation of Spinal Architecture

A spinal T10 contusion injury was created in Sprague-Dawley rats (*n*=52) with a Precision Systems IH Impactor (220 kDyn). Scaffolds (1.0 mm diameter, 2.0 mm length) were surgically implanted at the lesion site between 24 and 72 hours later. Rats were sacrificed at 12 weeks and histomorphometric analysis was performed on H&E stained sections.

Neuro-Spinal Scaffold Design

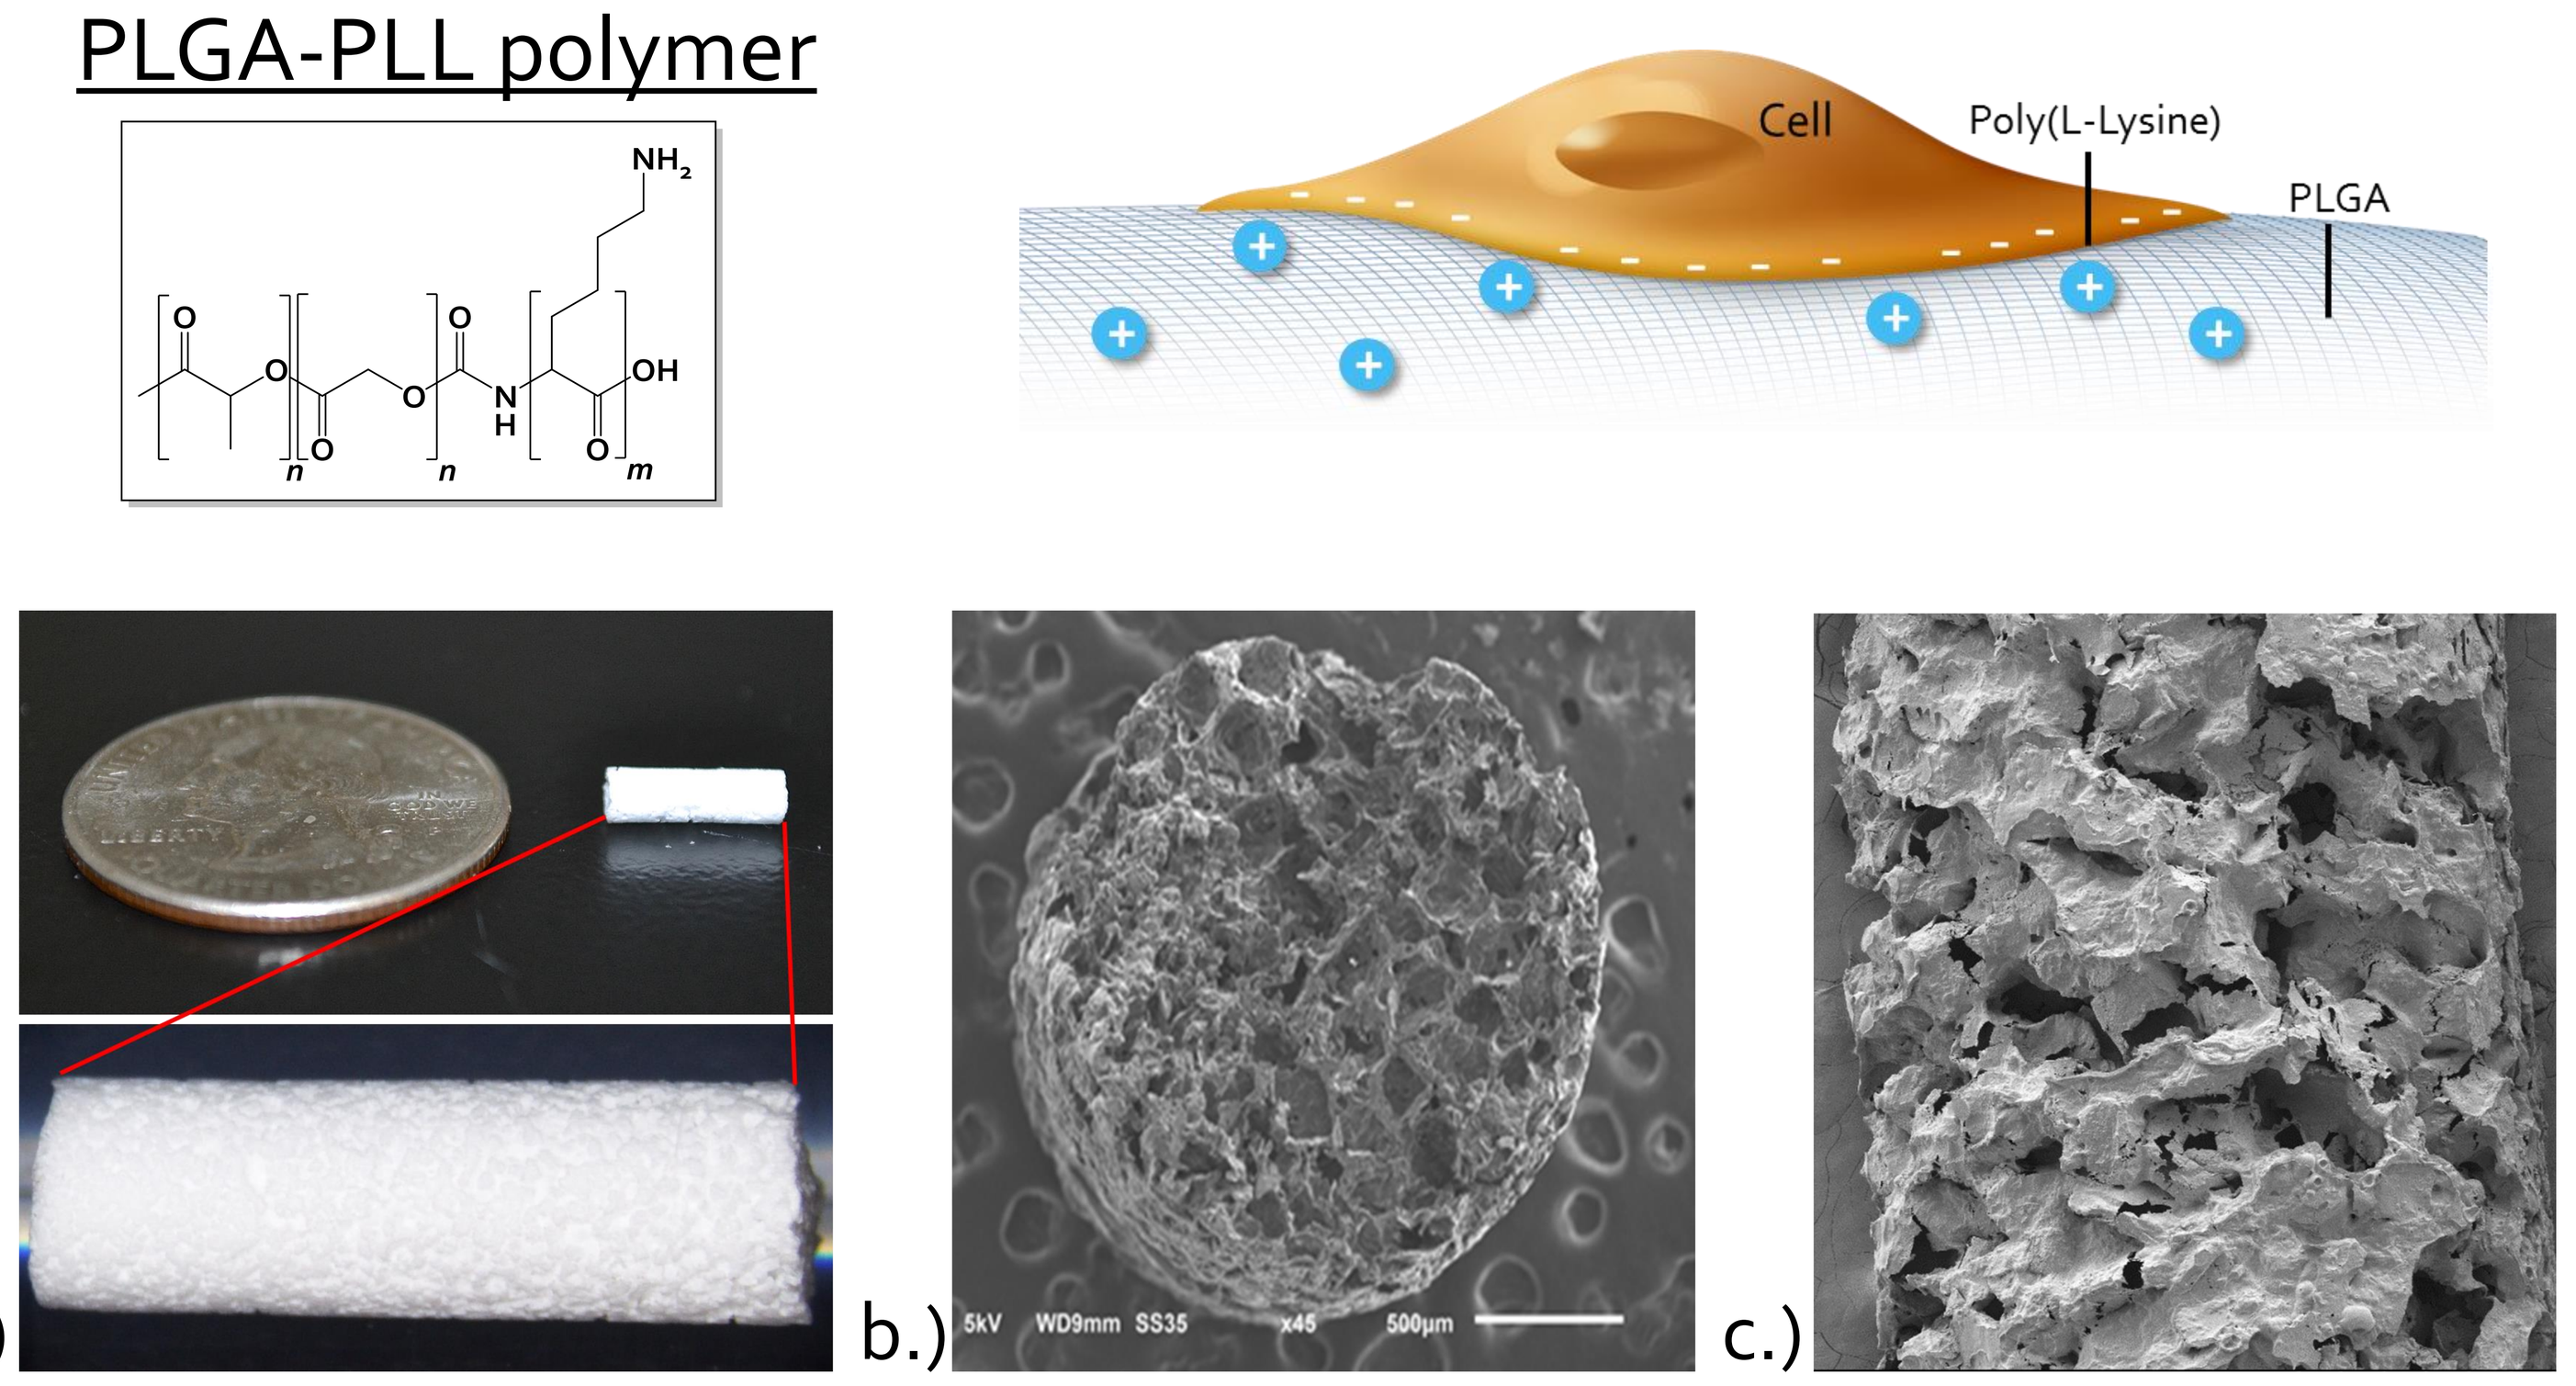
PLGA-PLL polymer
NH2
OH
Cell
Poly(L-Lysine)
PLGA
a.)
b.)
c.)

Figure 1: Images of a porous, cylindrical Neuro-Spinal Scaffold. a.) Macroscopic view of a 3mm (diameter) x 10mm (length) device b.) SEM of cross-sectional porosity c.) SEM of circumferential porosity.

Pre-Clinical Results

Surgical Feasibility of Neuro-Spinal Scaffold Implantation in a Porcine Model of Acute Contusion Spinal Cord Injury

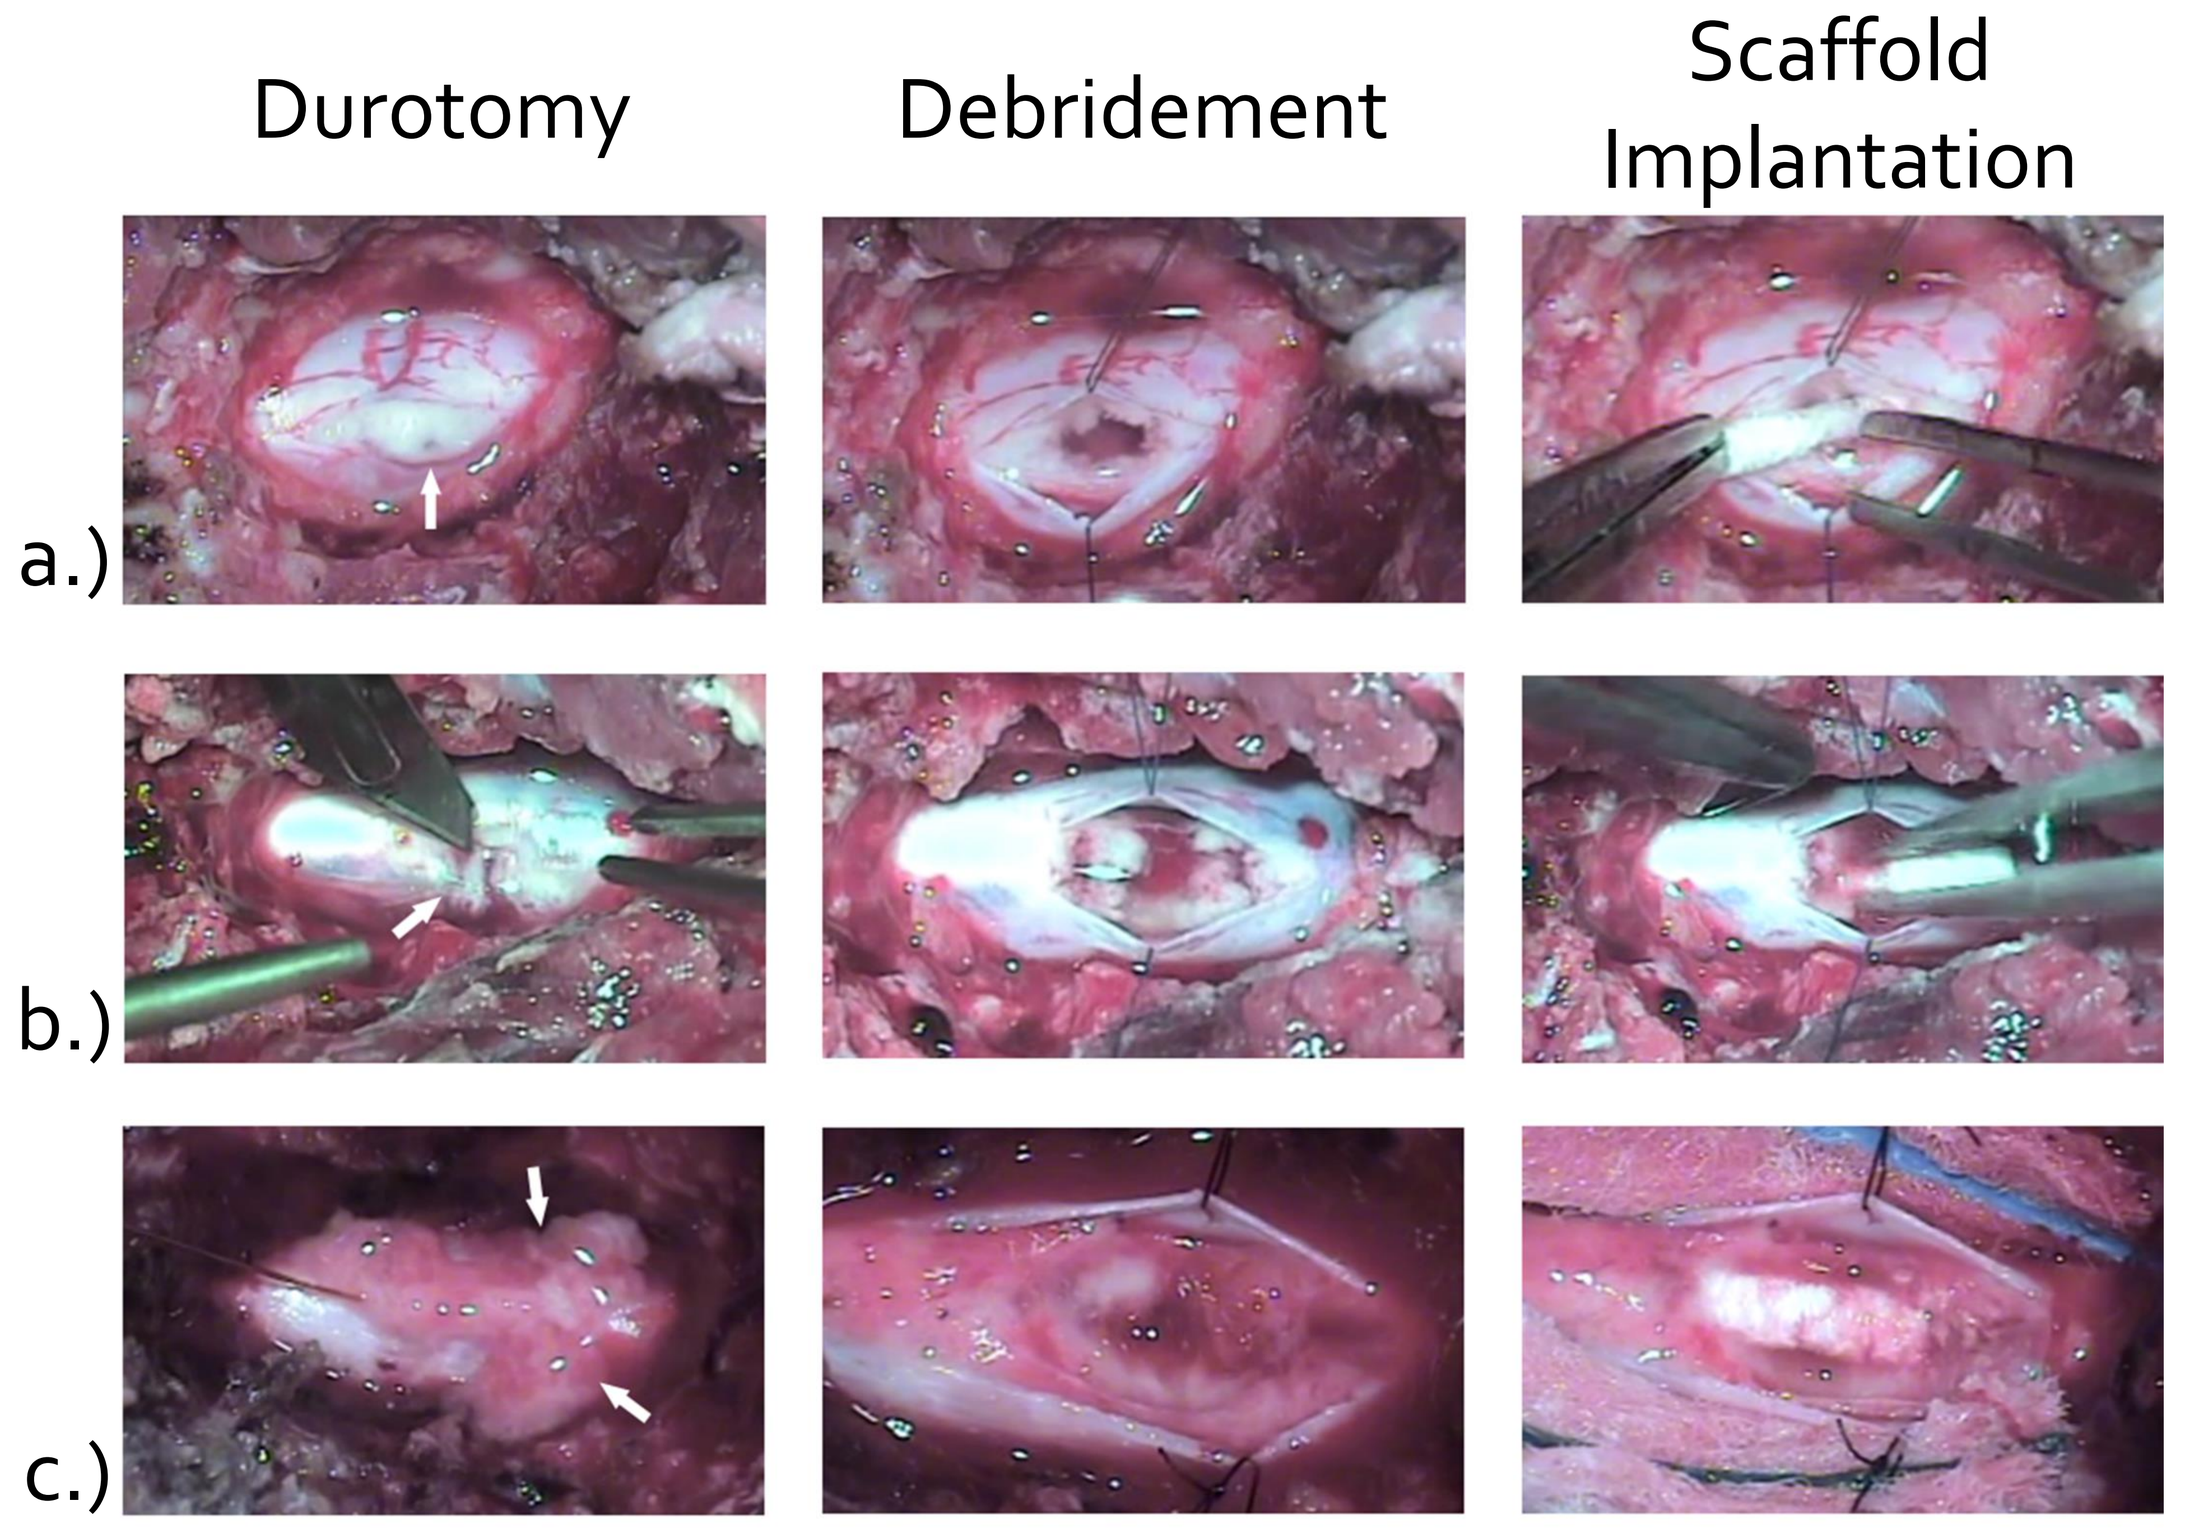
Durotomy
Debridement
Scaffold Implantation
a.)
b.)
c.)

Figure 2: Intraoperative images following durotomy (first column), saline irrigation illustrating cavitation (second column), and scaffold implantation (third column) at a.) 4 hrs b.) 6 hrs and c.) 24 hrs post contusion injury. White arrows in the first column highlight the spontaneous evacuation of necrotic neural tissue.

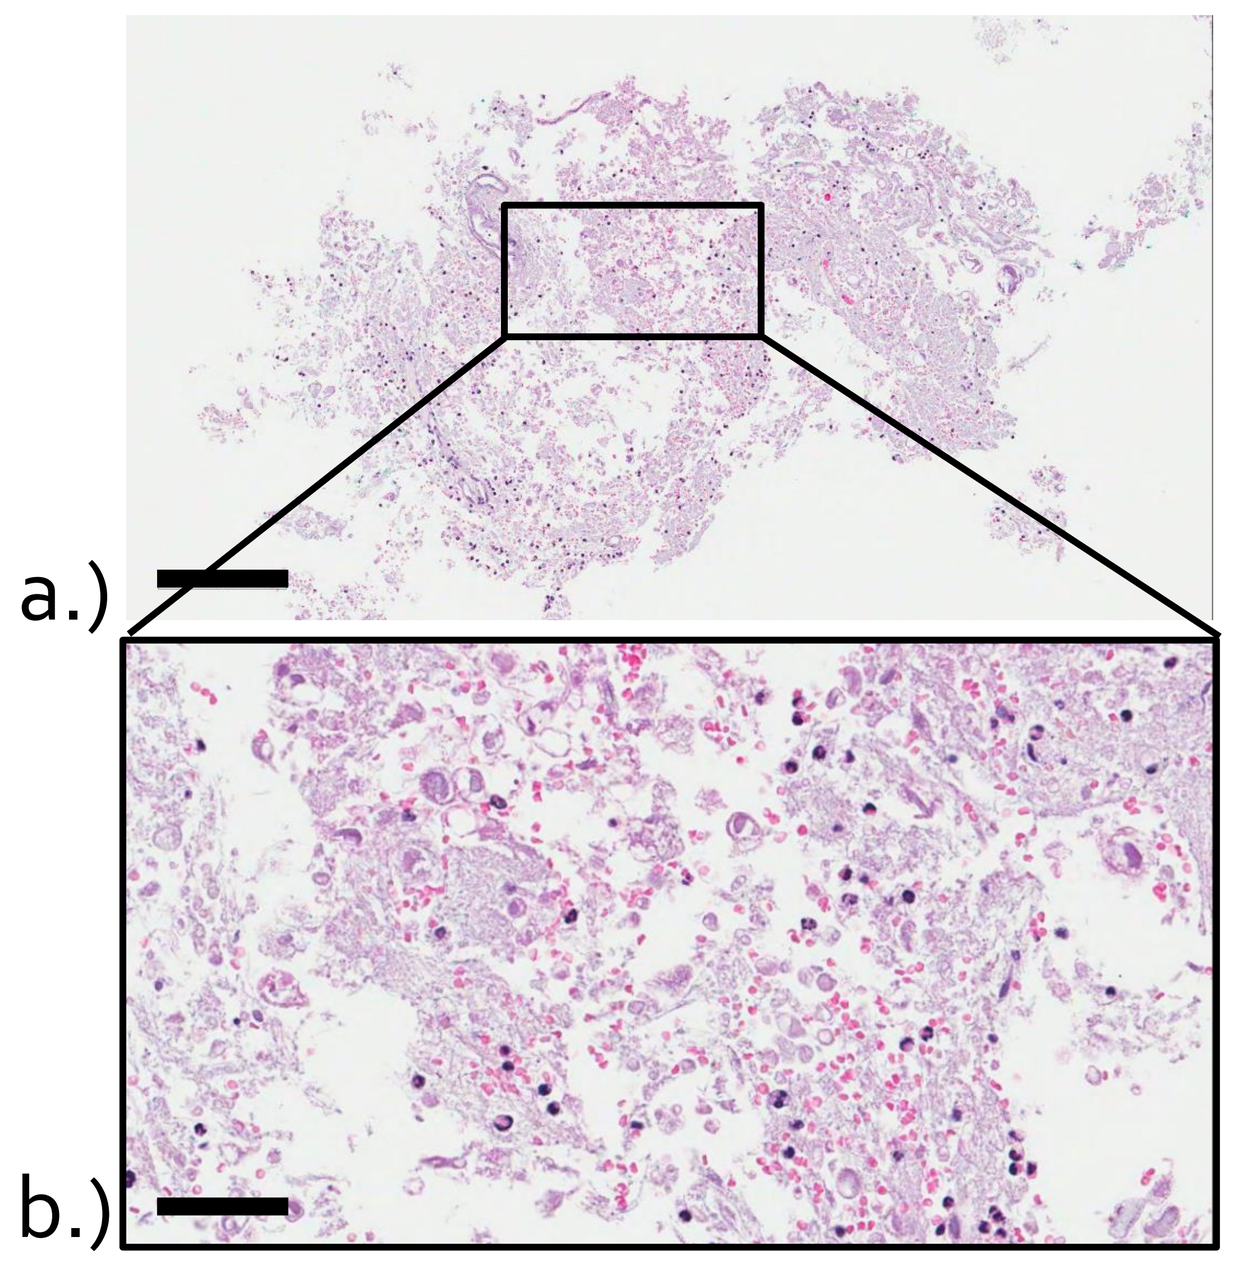
a.)
b.)

Figure 3: Histology (H&E) of collected necrotic neural tissue a.) 24 hrs (10x magnification), and b.) 24 hrs (40x magnification of black box in a.) post-injury. Scale bars represent 200 µm (a) and 50 µm (b).

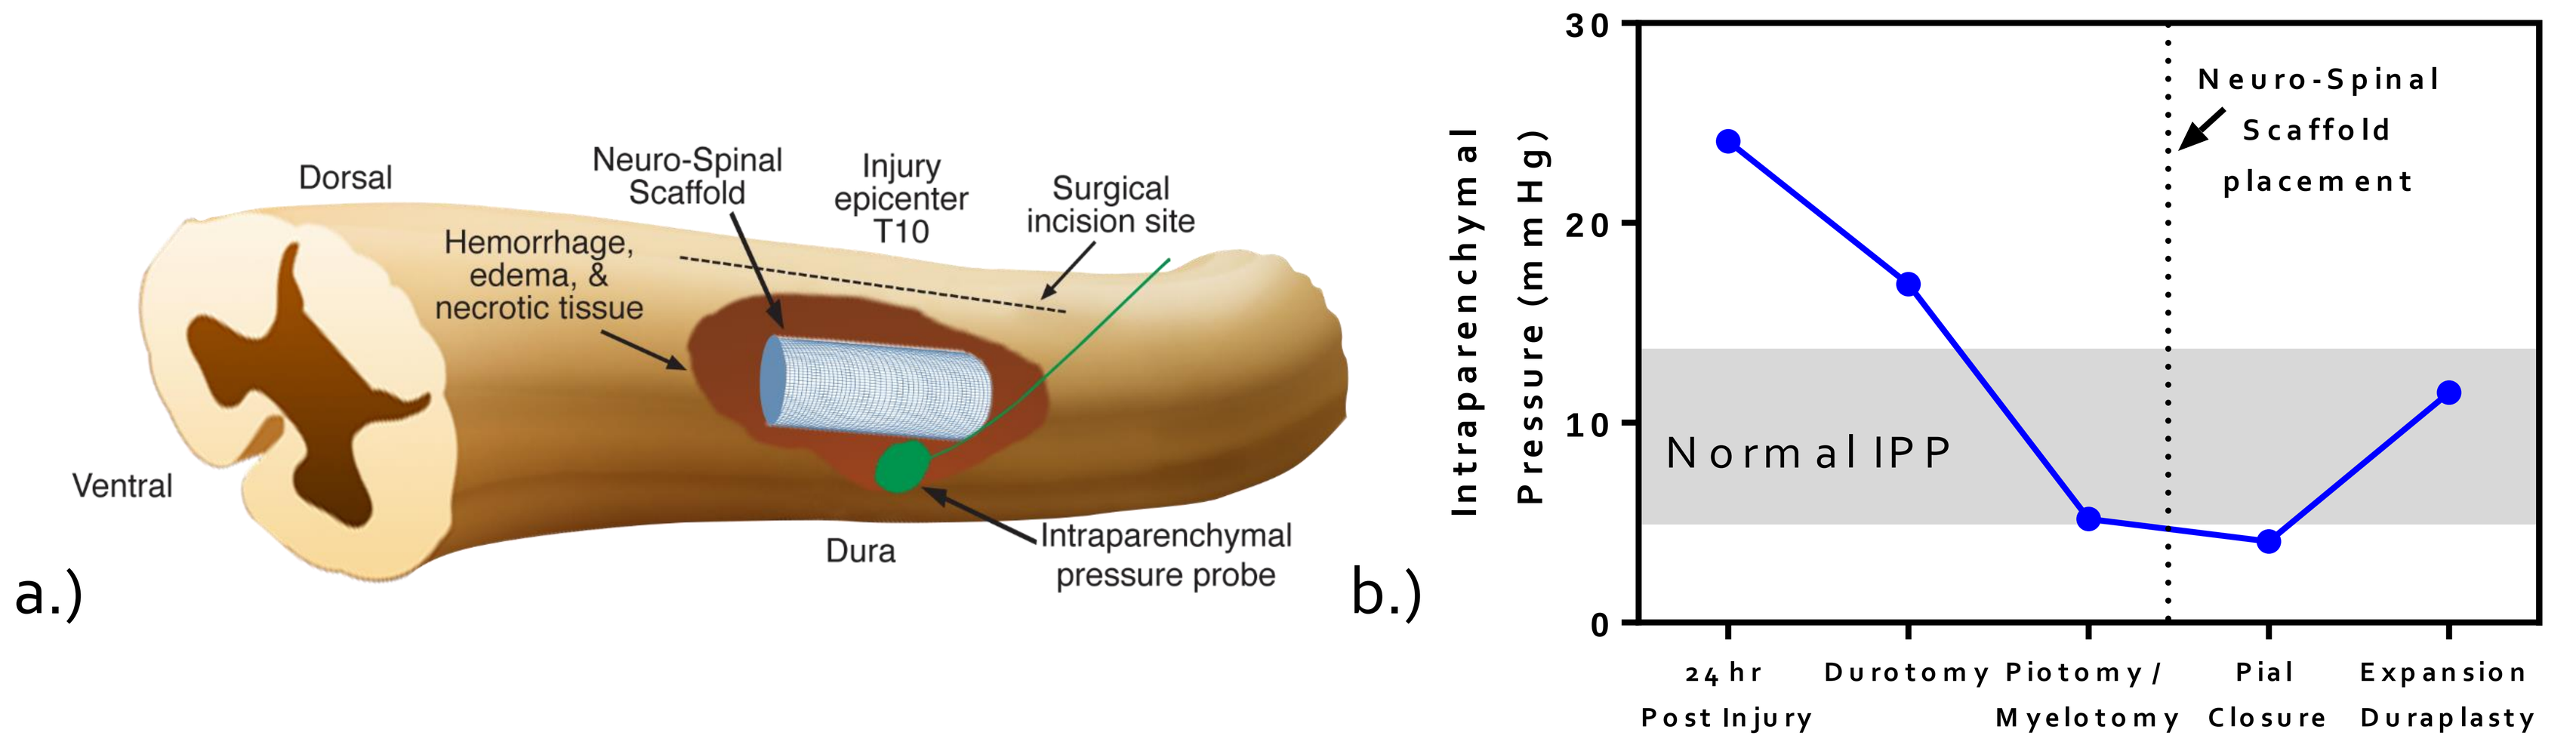
Dorsal
Neuro-Spinal Scaffold
Injury epicenter T10
Surgical incision site
Hemorrhage, edema, & necrotic tissue
Ventral
Dura
Intraparenchymal pressure probe
a.)
b.)
Intraparenchymal Pressure (mmHg)
Neuro-Spinal Scaffold placement
Normal IPP
24 hr Post Injury
Durotomy
Piotomy / Myelotomy
Pial Closure
Expansion Duraplasty

Figure 4: Acute intraparenchymal pressure monitoring during scaffold implantation. a.) Schematic of pressure measurement methodology b.) Pressure measurements following discrete surgical steps related to scaffold implantation at 24 hrs post contusion. A 1F pressure probe was inserted within 2cm of the injury site for all measurements. Data points calculated from continuous data averaged over one minute.

The Neuro-Spinal Scaffold Acts as a Physical Substrate to Preserve Spinal Cord Architecture in a Rat Contusion Model

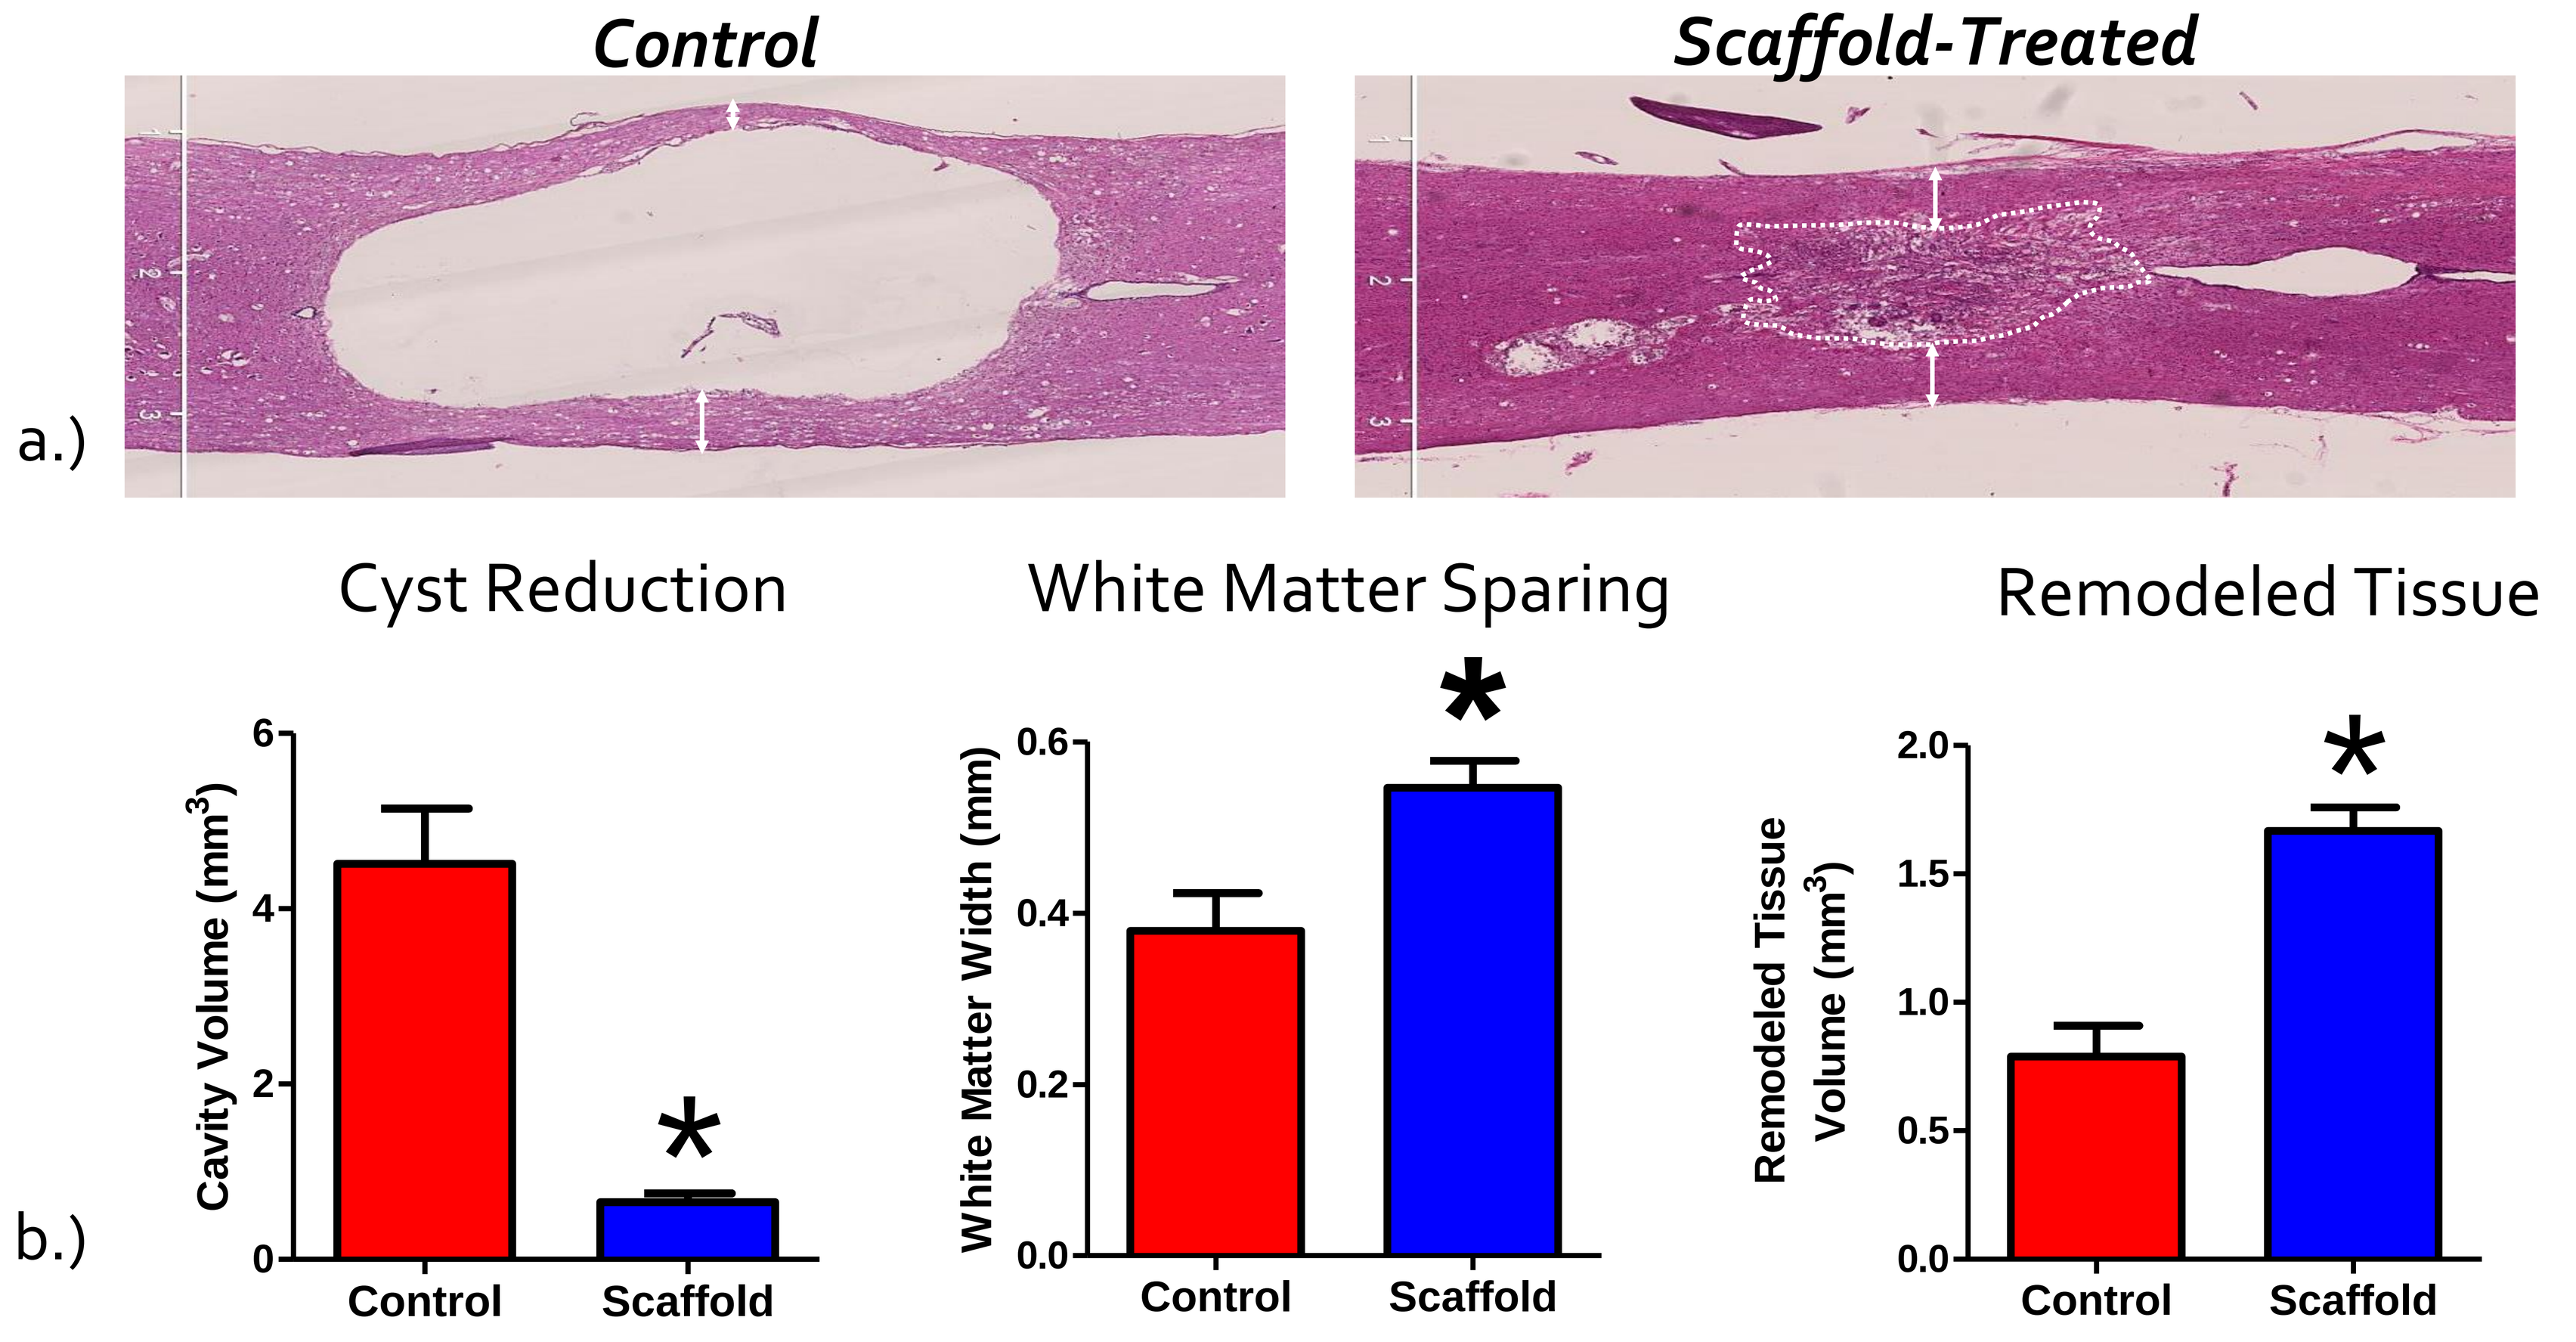
Control
Scaffold-Treated
a.)
Cyst Reduction
White Matter Sparing
Remodeled Tissue
b.)
Cavity Volume (mm³)
White Matter Width (mm)
Remodeled Tissue Volume (mm³)
Control
Scaffold
*

Figure 5: Neuro-Spinal Scaffold preserves spinal architecture. a.) Representative longitudinal sections from control (n=14) and scaffold implanted rats (n=38), b.) histomorphometric analysis (values are means ± S.E.M, *P<0.05).

Pilot Clinical Trial

Pilot Study of Clinical Safety and Feasibility of the PLGA Poly-L-Lysine Scaffold for the Treatment of Complete (AIS A) Traumatic Acute Spinal Cord Injury (NCT02138110)

The purpose of this study is to evaluate whether the scaffold is safe and feasible for the treatment of complete functional SCI and gather exploratory evidence of clinical effectiveness.

Key Inclusion Criteria:

• *AIS A (T3-T12/L1)* • *18-65 years of age* • *Non-penetrating contusion injury* • *< 4 days since injury* •

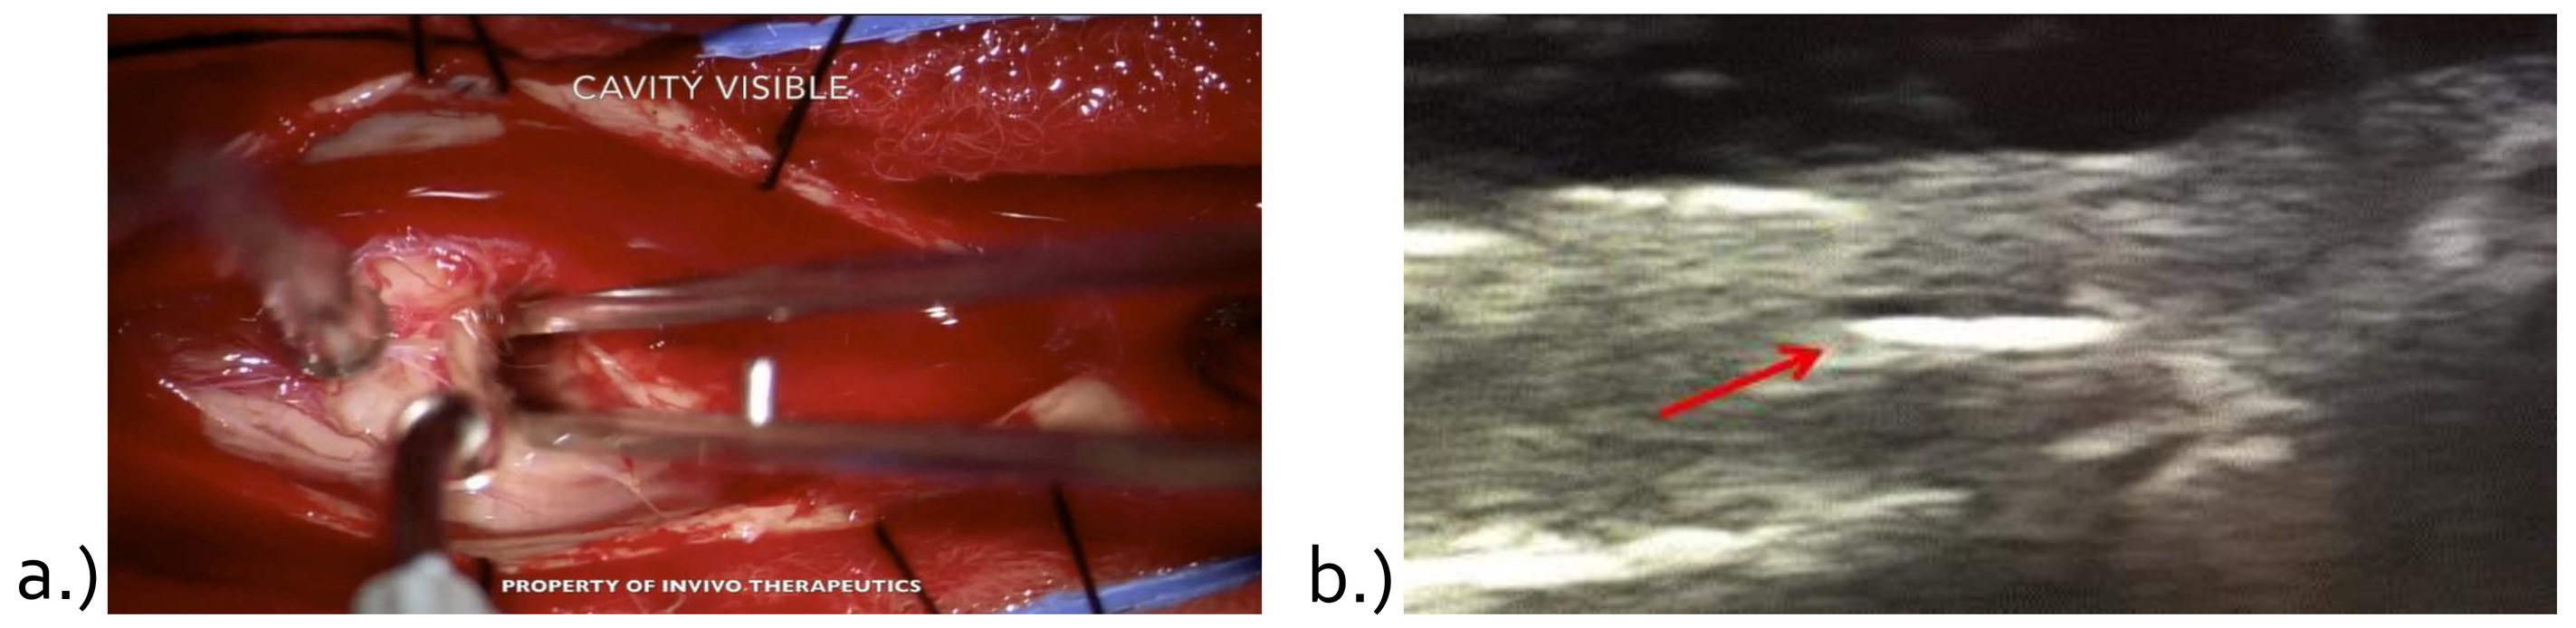
CAVITY VISIBLE
PROPERTY OF INVIVO THERAPEUTICS
a.)
b.)

Figure 6: First-in-human surgical implantation of Neuro-Spinal Scaffold. a.) Intraoperative image of acute cavity allowing for ease of scaffold implantation b.) Intraoperative ultrasound following scaffold implantation.

Clinical Update

- 4 of 5 Pilot subjects enrolled to date
- Subject 1 (T11) converted to AIS C at 1 month post-injury and demonstrated continued LEMS improvement at 6 months
- Subject 2 (T7) remains AIS A with marked sensory improvement
- Subject 3 (T4) converted to AIS B at 1 month post-injury
- Subject 4 (T3) was enrolled/implanted in August 2015
- No adverse events related to the Neuro-Spinal Scaffold
- No serious adverse events associated with the surgical procedure

Conclusion

- Acute necrosis and spinal cord cavitation occurs rapidly following contusion injury in both pre-clinical models and human injury allowing for ease of Neuro-Spinal Scaffold implantation.
- Surgical implantation of the Neuro-Spinal Scaffold results in intraparenchymal pressure normalization in a porcine contusion model.
- Implantation of the Neuro-Spinal Scaffold preserves the spinal cord architecture in a rat contusion model which may provide a neuropermissive environment for neural regeneration.
- Preliminary clinical findings are promising, however further investigation is required to elucidate and confirm the therapeutic effect.

References

1. Teng YD, Lavik EB, Qu X, Park KI, Ourednik J, Zurakowski D, et al: Proc Natl Acad Sci U S A 99:3024-3029, 2002
2. Pritchard CD, Slotkin JR, Yu D, Dai H, Lawrence MS, Bronson RT, et al: J Neurosci Methods 188:258-269, 2010.
3. Bertram JP, Jay SM, Hynes SR, Robinson R, Criscione JM, Lavik EB: Acta Biomater 5:2860-2871, 2009.
4. Lee JH, Jones CF, Okon EB, Anderson L, Tigchelaar S, Kooner P, et al: A novel porcine model of traumatic thoracic spinal cord injury. J Neurotrauma 30:142-159, 2013.